Certification of Carbon Monoxide Diffusing Capacity Measurements for Clinical Trials of Inhaled Insulin

R.L. Jensen, PhD, R.O. Crapo, MD, P-Y Berclaz. Muchmore, MD, Salt Lake City, UT, Indianapolis, IN

ABSTRACT

CERTIFICATION OF CARBON MONOXIDE DIFFUSING CAPACITY MEASUREMENTS FOR CLINICAL TRIALS OF INHALED INSULIN.

RL Jensen, RO Crapo, P-Y Berclaz, C Zimmer, D Muchmore. LDS Hospital, Salt Lake City, UT, Eli Lilly and Co., Indianapolis, IN, RDS Boulder, CO.

Clinical trials of AIR® Inhaled Insulin (AIR® is a registered trademark of Alkermes, Inc) include serial measurements of carbon monoxide diffusing capacity (DLco) as one of several safety parameters. The measurements are performed in the United States and other countries, in widely different laboratory settings, using 12 different pulmonary function-testing (PFT) systems. To assure quality, laboratories follow standard calibration and testing methods outlined by the American Thoracic Society (ATS). Before testing patients in a clinical trial, laboratories use a DLco simulator (Hans Rudolph, Kansas City, MO) to validate and monitor the accuracy of their DLco measurements. The DLco simulator delivers a known inspired volume and precise, known "exhaled" gas concentrations. When combined with the breath hold time from the PFT device, exact “target” values for DLco can be calculated. Simulations with four different target values are introduced into each device by varying exhaled gas concentrations and inspired volumes. Differences between measured and target values for alveolar volume (VA), inspired volume and gas concentrations of carbon monoxide and the tracer gas are then evaluated. A DLco device is certified to be functioning properly if measured volumes are within 3% of target and DLco measures are within 3.0 standard DLco units (ml CO/min/mmHg) of calculated target values. After initial certification, simulation testing is done each week for three months and then bi-weekly for the remainder of the study.

Results of the initial DLco simulation tests from 125 pulmonary laboratories	
94 (75.2%)	Passed. Many sites required coaching on technique but no hardware
24 (19.2%)	Failed. Passed after servicing and/or replacing the test gas
6 (4.8%)	Failed. Passed after purchasing new equipment
1 (0.8%)	Failed. Unable to certify equipment - site dropped

Equipment at thirty-one sites (24.8%) failed the initial DLco simulation testing. After correcting instrument issues or purchasing new equipment, 124 (99.2 %) sites were confirmed to be measuring DLco accurately. A significant percentage of the DLco devices in normal clinical pulmonary function labs are

Introduction /Methods

Introduction

Large scale clinical trials have been implemented to determine safety for inhaled insulin. To assess safety in globally randomized controlled trials, primary outcome measurements of forced vital capacity (FVC), forced exhaled volume in one second (FEV1) and carbon monoxide diffusing capacity (DLco) are measured . Laboratory control for FVC and FEV1 are well established, however determining accuracy of laboratory measured DLco has been difficult. A DLco simulator (Hans Rudolph, Kansas City, MO) capable of injecting a known, repeatable DLco value into any DLco measuring device. These simulators can easily quality assess DLco devices on a regular basis within each laboratory during these large scale global clinical trials.

The DLco simulator is comprised of two syringes (3.0 and 5.0 liter) that are joined by a manifold that directs the flow into and out of one or the other of the two syringes. The basic principle of the simulator is that it mimics an actual patient DLco test (pre-breathing, inhalation, breath-hold and exhalation). The pre-breathing and inhalation phases use the 5.0 liter syringe which can be set by use of a collar to inhale an exact volume of DLco test gas. During the breath-hold the time the operator turns the valve on the manifold to redirect the DLco simulator to the 3.0 liter syringe. Prior to starting the simulation test, this syringe is filled with a precision gas mixture that emulates typical alveolar gas concentration seen in patients. After the breath-hold the content of the 3.0 liter syringe is emptied into the DLco device under test.

Software developed for the DLco simulator is used to calculate precise target DLco, VA and IVC and differences between these and the actual measured values. Test results are then reported and stored in a database.

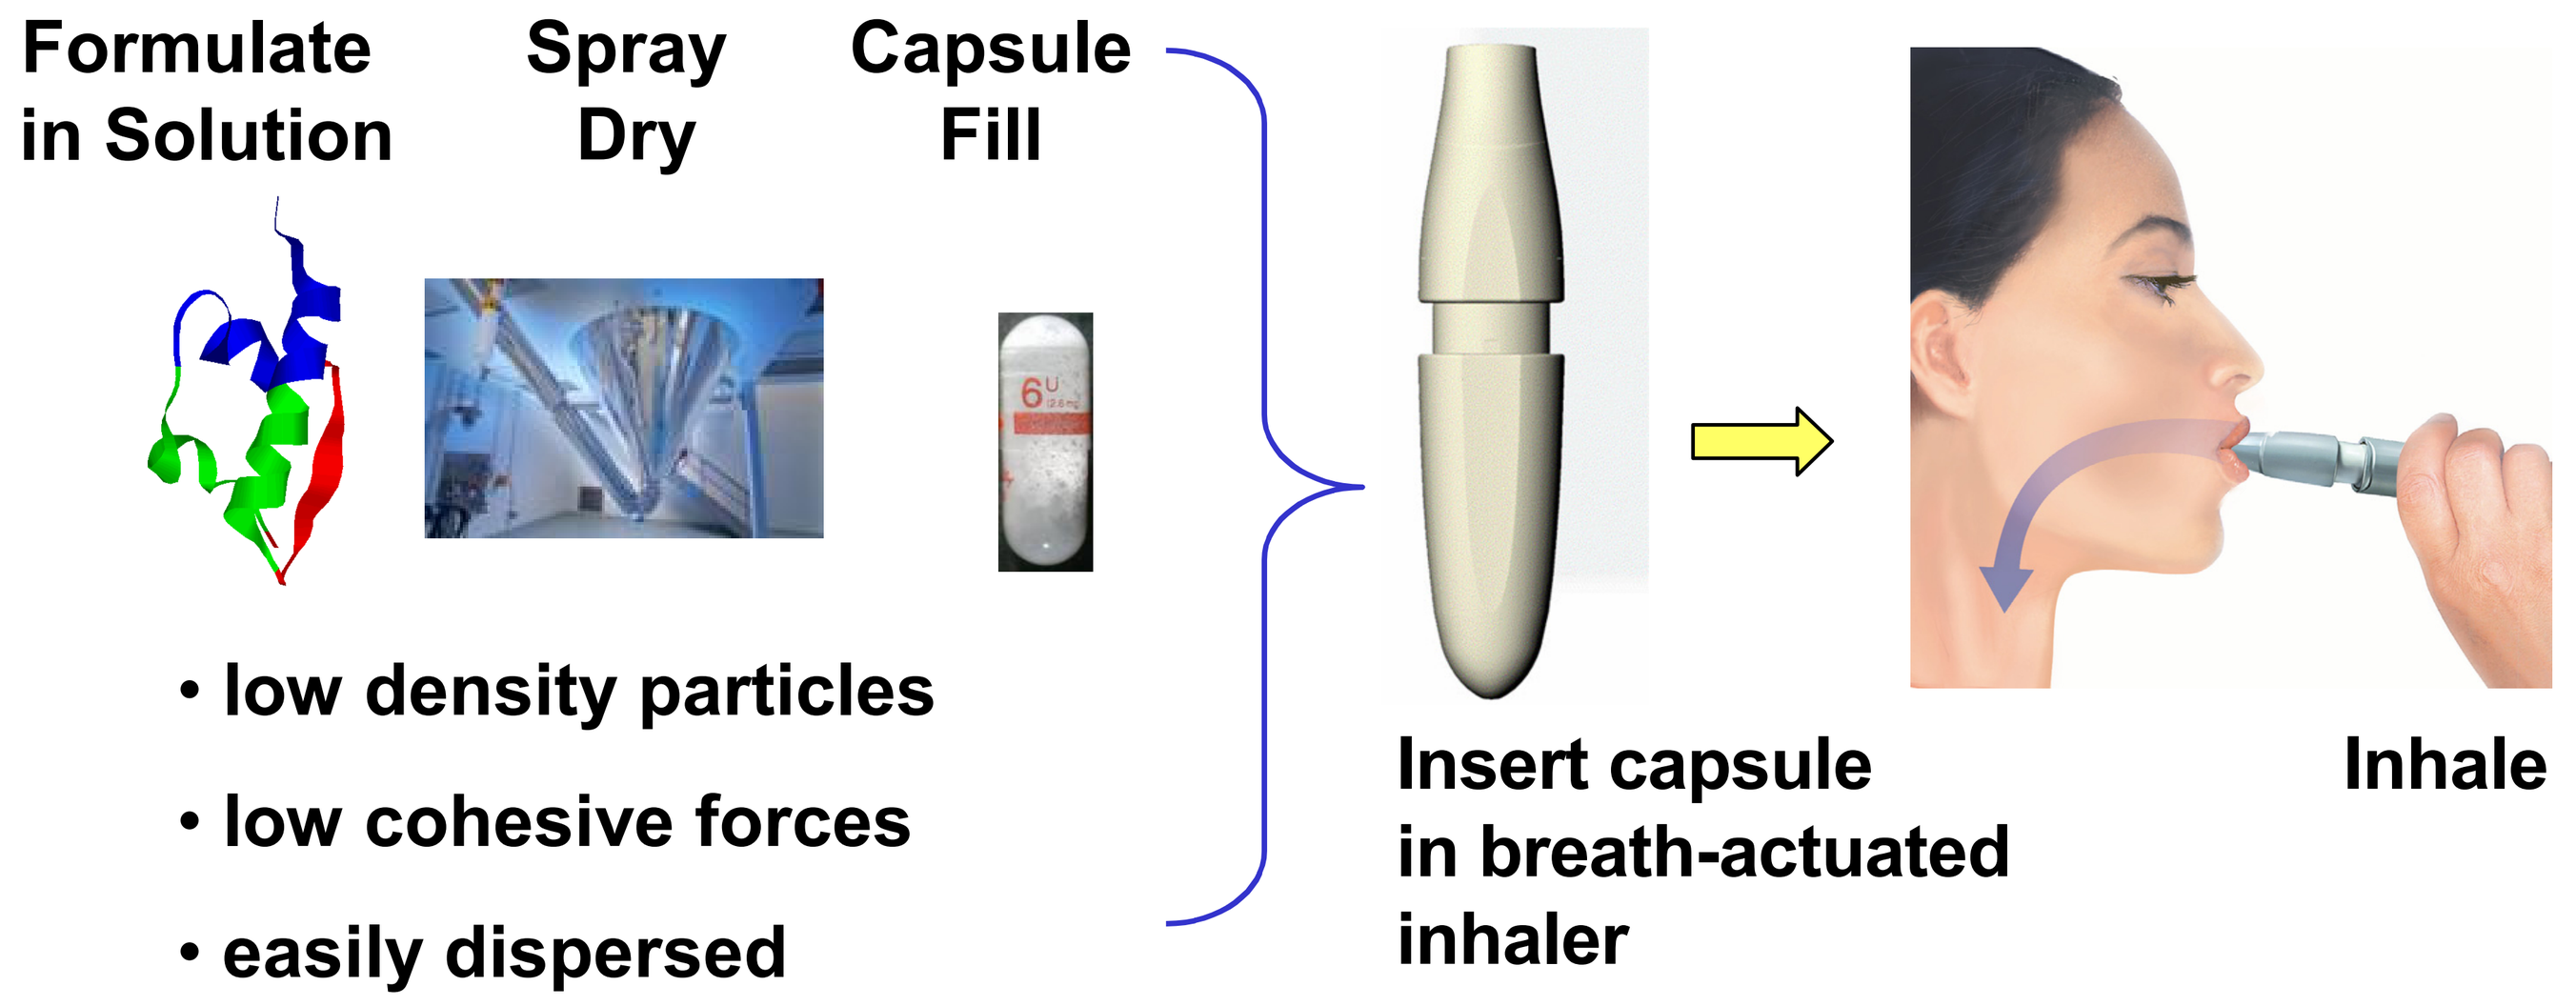
Formulate in Solution
Spray Dry
Capsule Fill
6U
• low density particles
• low cohesive forces
• easily dispersed
Insert capsule in breath-actuated inhaler
Inhale

ANALYSIS

Methods

<u>Laboratory Selection and Patient Testing</u>

- 125 existing hospital or clinic PFT labs recruited to participate
- Prior to the study, all labs submitted PFTs from 10 different subjects for review by the authors (RJ or ROC) to assess compliance with ATS standards and the lab’s capability to perform the approved protocol
- PFT technicians were trained and certified at a centralized meeting
- Measurements of FVC, FEV_1, diffusing capacity (DL_{co}), and total lung capacity (TLC) were performed on each subject

<u>DLco Simulator Testing</u>

- Before randomizing patients into the clinical trials, laboratories are required to use a DLco simulator (Hans Rudolph, Kansas City, MO) to validate and monitor the accuracy of their DLco measurements.
- The measurements are performed in the United States and other countries, in widely different laboratory settings, using at least 12 different pulmonary function-testing (PFT) systems.
- Even among similar PFT systems there were model and software differences.
- A DLco device is certified to be functioning properly if measured volumes are within 3% of target and DLco measures are within 3.0 standard DLco units (ml CO/min/mmHg) of calculated target values.

RESULTS/CONCLUSIONS

Results

Figure 1 below shows differences between DLco simulation target and measured values vs. simulator target.

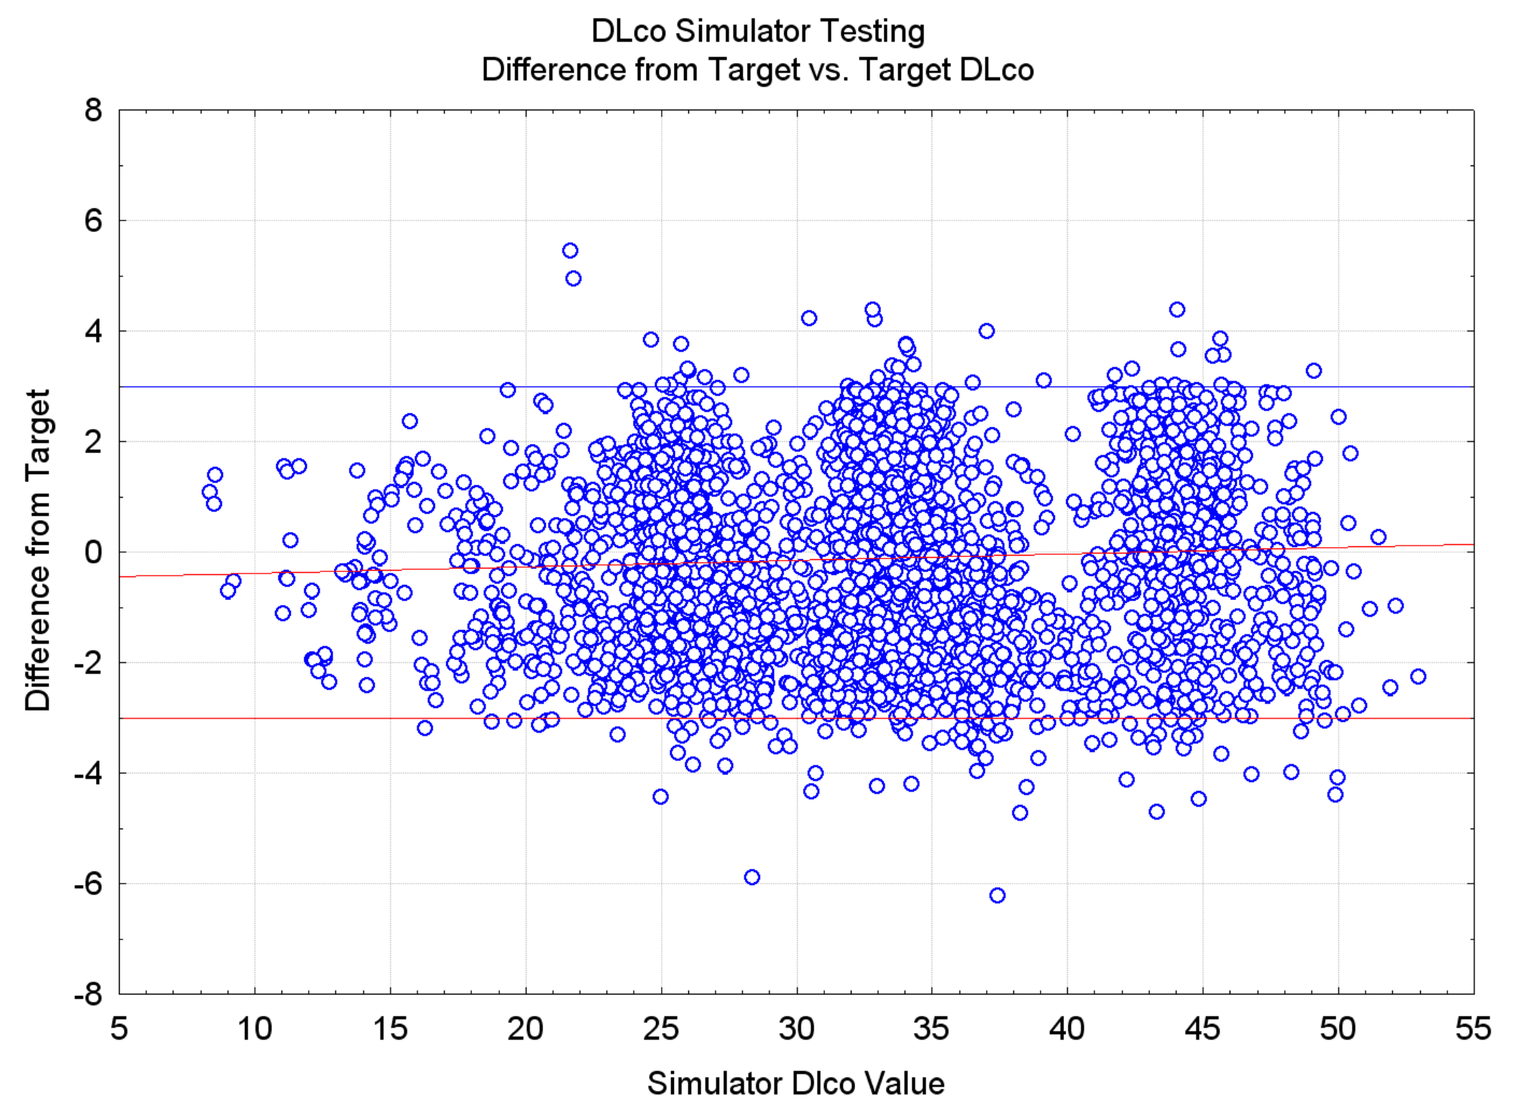
DLco Simulator Testing
Difference from Target vs. Target DLco
Difference from Target
Simulator Dlco Value

Figure 2 below shows differences between DLco simulation target and measured values.

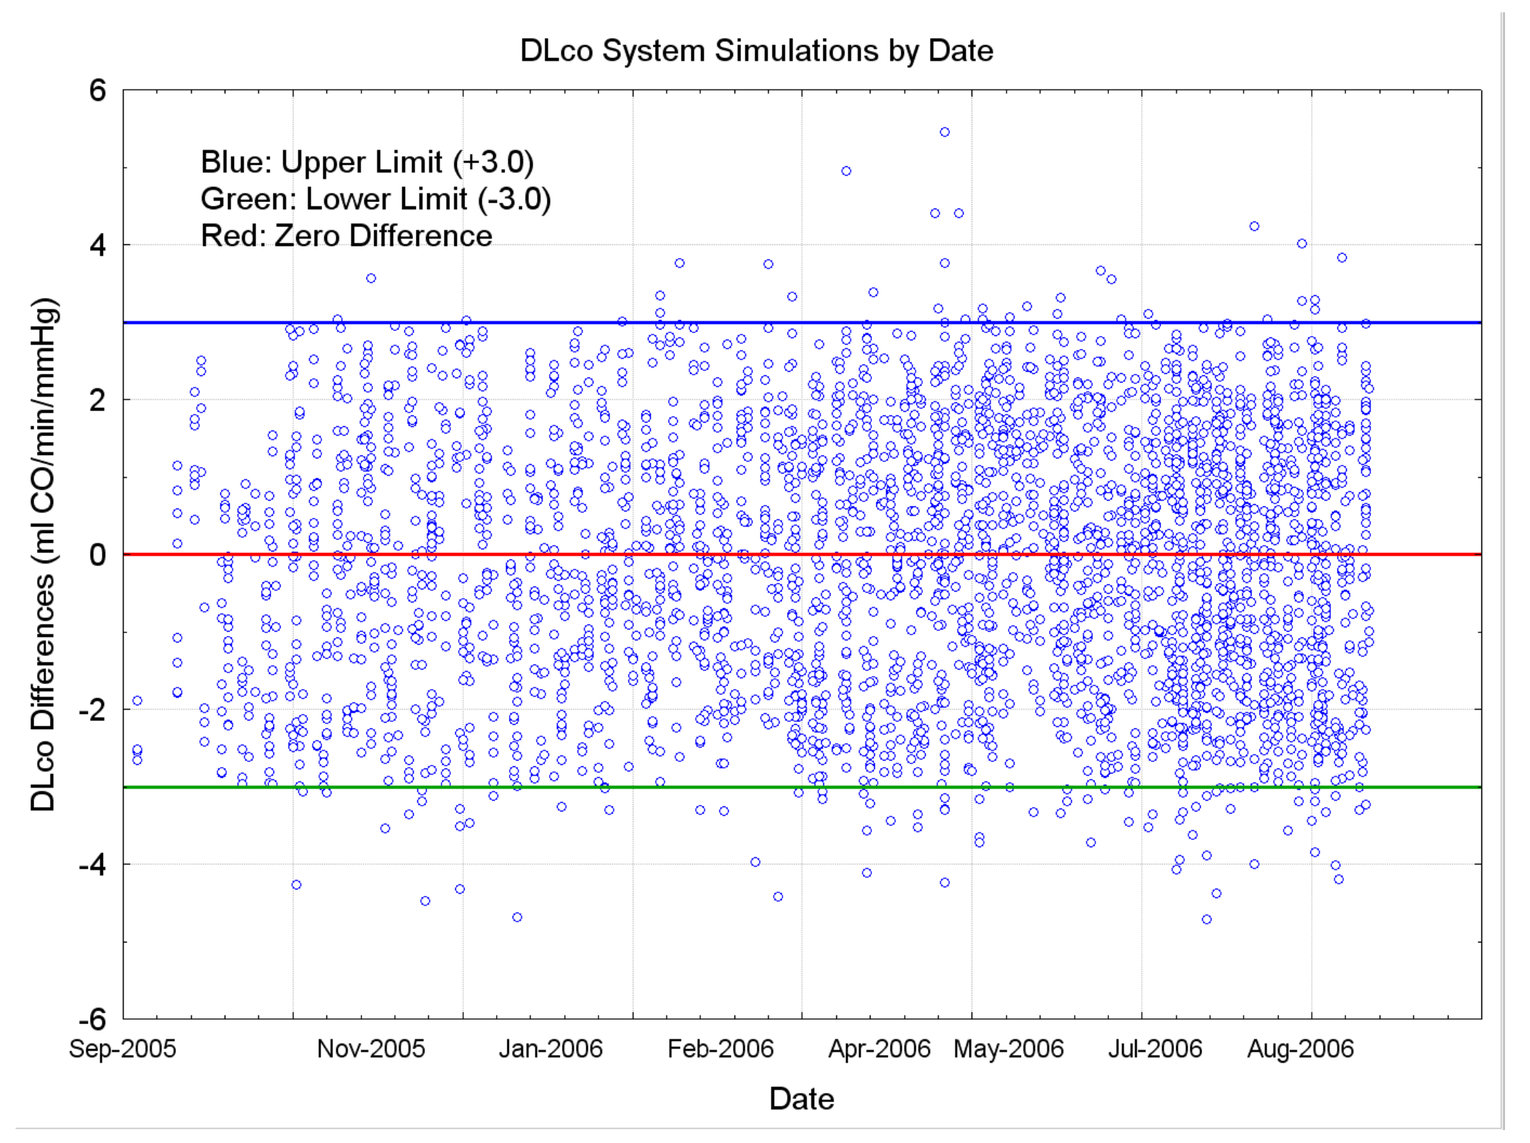
DLco System Simulations by Date
Blue: Upper Limit (+3.0)
Green: Lower Limit (-3.0)
Red: Zero Difference
DLco Differences (ml CO/min/mmHg)
Sep-2005
Nov-2005
Jan-2006
Feb-2006
Apr-2006
May-2006
Jul-2006
Aug-2006
Date

Figure 2 below shows differences between DLco simulation target and measured values.

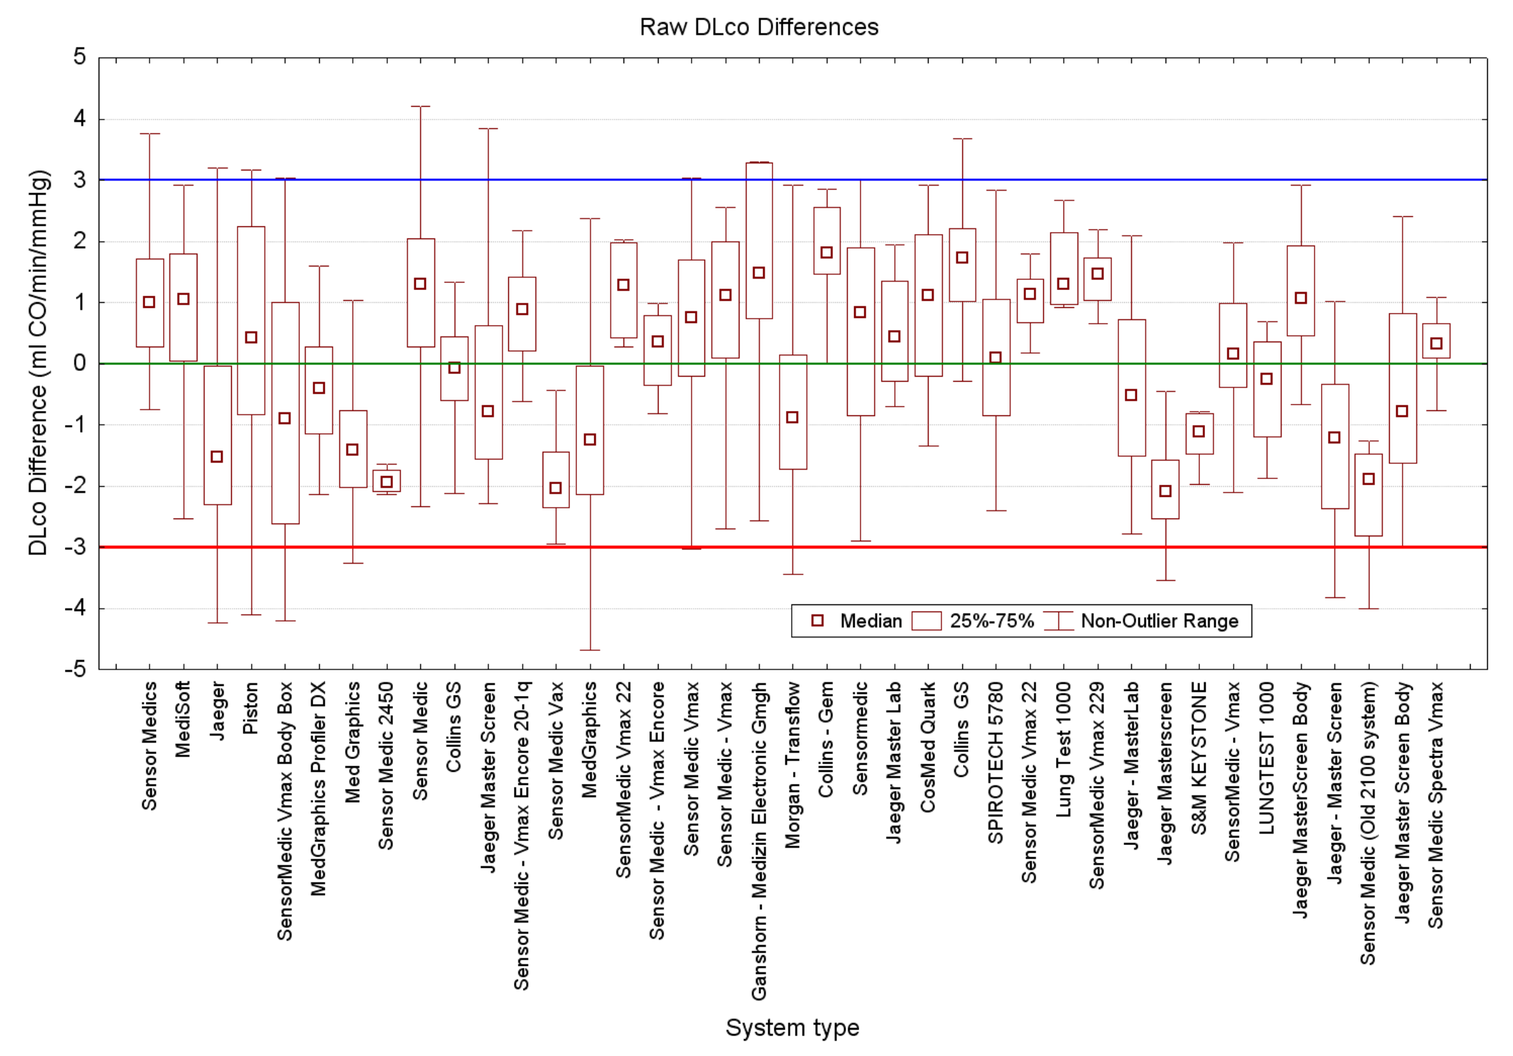
Raw DLco Differences
DLco Difference (ml CO/min/mmHg)
Median
25%-75%
Non-Outlier Range
System type

Results of the initial DLco simulation tests from 125 pulmonary laboratories

94 (75.2%)	Passed. However, many sites required coaching on technique but no hardware issues were found
24 (19.2%)	Initially failed. Later passed after servicing and/or replacing the DLco test gas
6 (4.8%)	Initially failed. Passed only after purchasing new equipment
1 (0.8%)	Failed. Unable to certify equipment - site dropped

CONCLUSIONS

- A significant percentage of the DLco devices in clinical pulmonary function labs are likely to be malfunctioning.
- This study underscores the need to certify and continuously monitor DLco test quality in clinical trials and clinical laboratories.